FRI-0910: TERIPARATIDE ACCELERATES PROXIMAL HUMERUS FRACTURE CONSOLIDATION – THE *TERAFRAP* STUDY

Barmherzige Schwestern Krankenhaus Wien

Christian Muschitz[1], Judith Haschka[1], Georg Langs[2], Markus Holzer[2], Andreas Baierl[3], Christoph Pümpel[1], Zora Messner[1], Roland Kocijan[1], Xaver Feichtinger[4], Rainer Mittermayr[4], Jakob Schanda[4], Thomas Hausner[5], Jochen Schmidsfeld[6], Robert Wakolbinger[1], Christian Fialka[4,7], Wolfgang Schima[8], and Heinrich Resch[1,7]

1. St. Vincent Hospital – Medical Department II – VINFORCE; Academic Teaching Hospital of the Medical University of Vienna, Stumpergasse 13, 1060 Vienna
2. Medical University of Vienna, Department of Biomedical Imaging and Image-guided Therapy, Computational Imaging Research Lab, Währinger Gürtel 18-20, 1090 Vienna, Austria
3. University of Vienna, Department of Statistics and Operations Research, Oskar-Morgenstern-Platz 1, 1090 Vienna, Austria
4. AUVA Trauma Center Meidling, Kundratstrasse 37, 1120 Vienna, Austria
5. AUVA Trauma Center Lorenz Böhler, Donaueschingenstraße 13, 1200 Vienna, Austria
6. Social Medicine Center East, Department of Traumatology, Langobardenstrasse 122, 1220 Vienna, Austria
7. Sigmund Freud University, Medical Faculty, Sigmund Freud Platz 1, 1020 Vienna, Austria
8. St. Vincent Hospital – Department of Diagnostic and Interventional Radiology; Academic Teaching Hospital of the Medical University of Vienna, Stumpergasse 13, 1060 Vienna, Austria

Purpose

Reviews of preclinical and clinical studies concluded that once-daily administration of systemic recombinant teriparatide [rhPTH (1-34), TPTD] enhanced the morphometric and mechanical properties of fracture calluses and accelerated fracture healing and consolidation suggested that this treatment may be used successfully in clinical practice especially at peripheral skeletal sites.

Systemic teriparatide ([rhPTH 1-34] TPTD) enhances callus formation and mechanical strength after fracture in animal models and in humans. Less data exist on three-dimensional (3D) structural effects at the proximal humerus, a specific non weight-bearing cortical and trabecular bone site with high fracture incidence.

Hypothesis and Objective

The hypothesis of this investigator initiated study was to test whether or not TPTD exerts positive effects on fracture consolidation in female and male patients with a recent humerus fracture.

The primary objective was the quantitative assessment of fracture consolidation in the treatment and placebo groups based on CT data and computational analysis of the bone structure. Bone structure was assessed by means of quantized bone texture characteristics in the vicinity of the fracture.

Inclusion criteria:

- recent proximal 2-segment humerus fracture (0 – 8 days post fracture)
- no surgical treatment at fractured site
- informed consent
- postmenopausal female and male patients aged 60 – 85 years
- established osteoporosis as defined by BMD measured with DXA-technology with a T-score ≤ -2.0 spine or hip

Exclusion criteria:

- any contraindication against teriparatide (TPTD, rhPTH 1-34)
- Hypersensitivity to the active substance or to any of the excipients.
- Pre-existing hypercalcemia
- Severe renal impairment (eGFR< 35ml/min)
- Metabolic bone diseases (including hyperparathyroidism and Paget's disease of the bone) other than primary osteoporosis or glucorticoid-induced osteoporosis.
- Unexplained elevations of alkaline phosphatase
- Prior external beam or implant radiation therapy to the skeleton
- Patients with skeletal malignancies or bone metastases should be excluded from treatment with teriparatide.
- any prior antiresorptive therapy (oral/intravenous bisphosphonates, RANKL-antibody, SERMs)
- any prior strontium ranelate therapy
- any prior TPTD of PTH 1-84 therapy
- malignancies ≤ 5 years except basalioma
- hypo-/hypercalcemia
- baseline 25-OH vitamin D3 level ≤10 ng/ml
- prosthesis at fractured and contralateral humerus

Figure 1

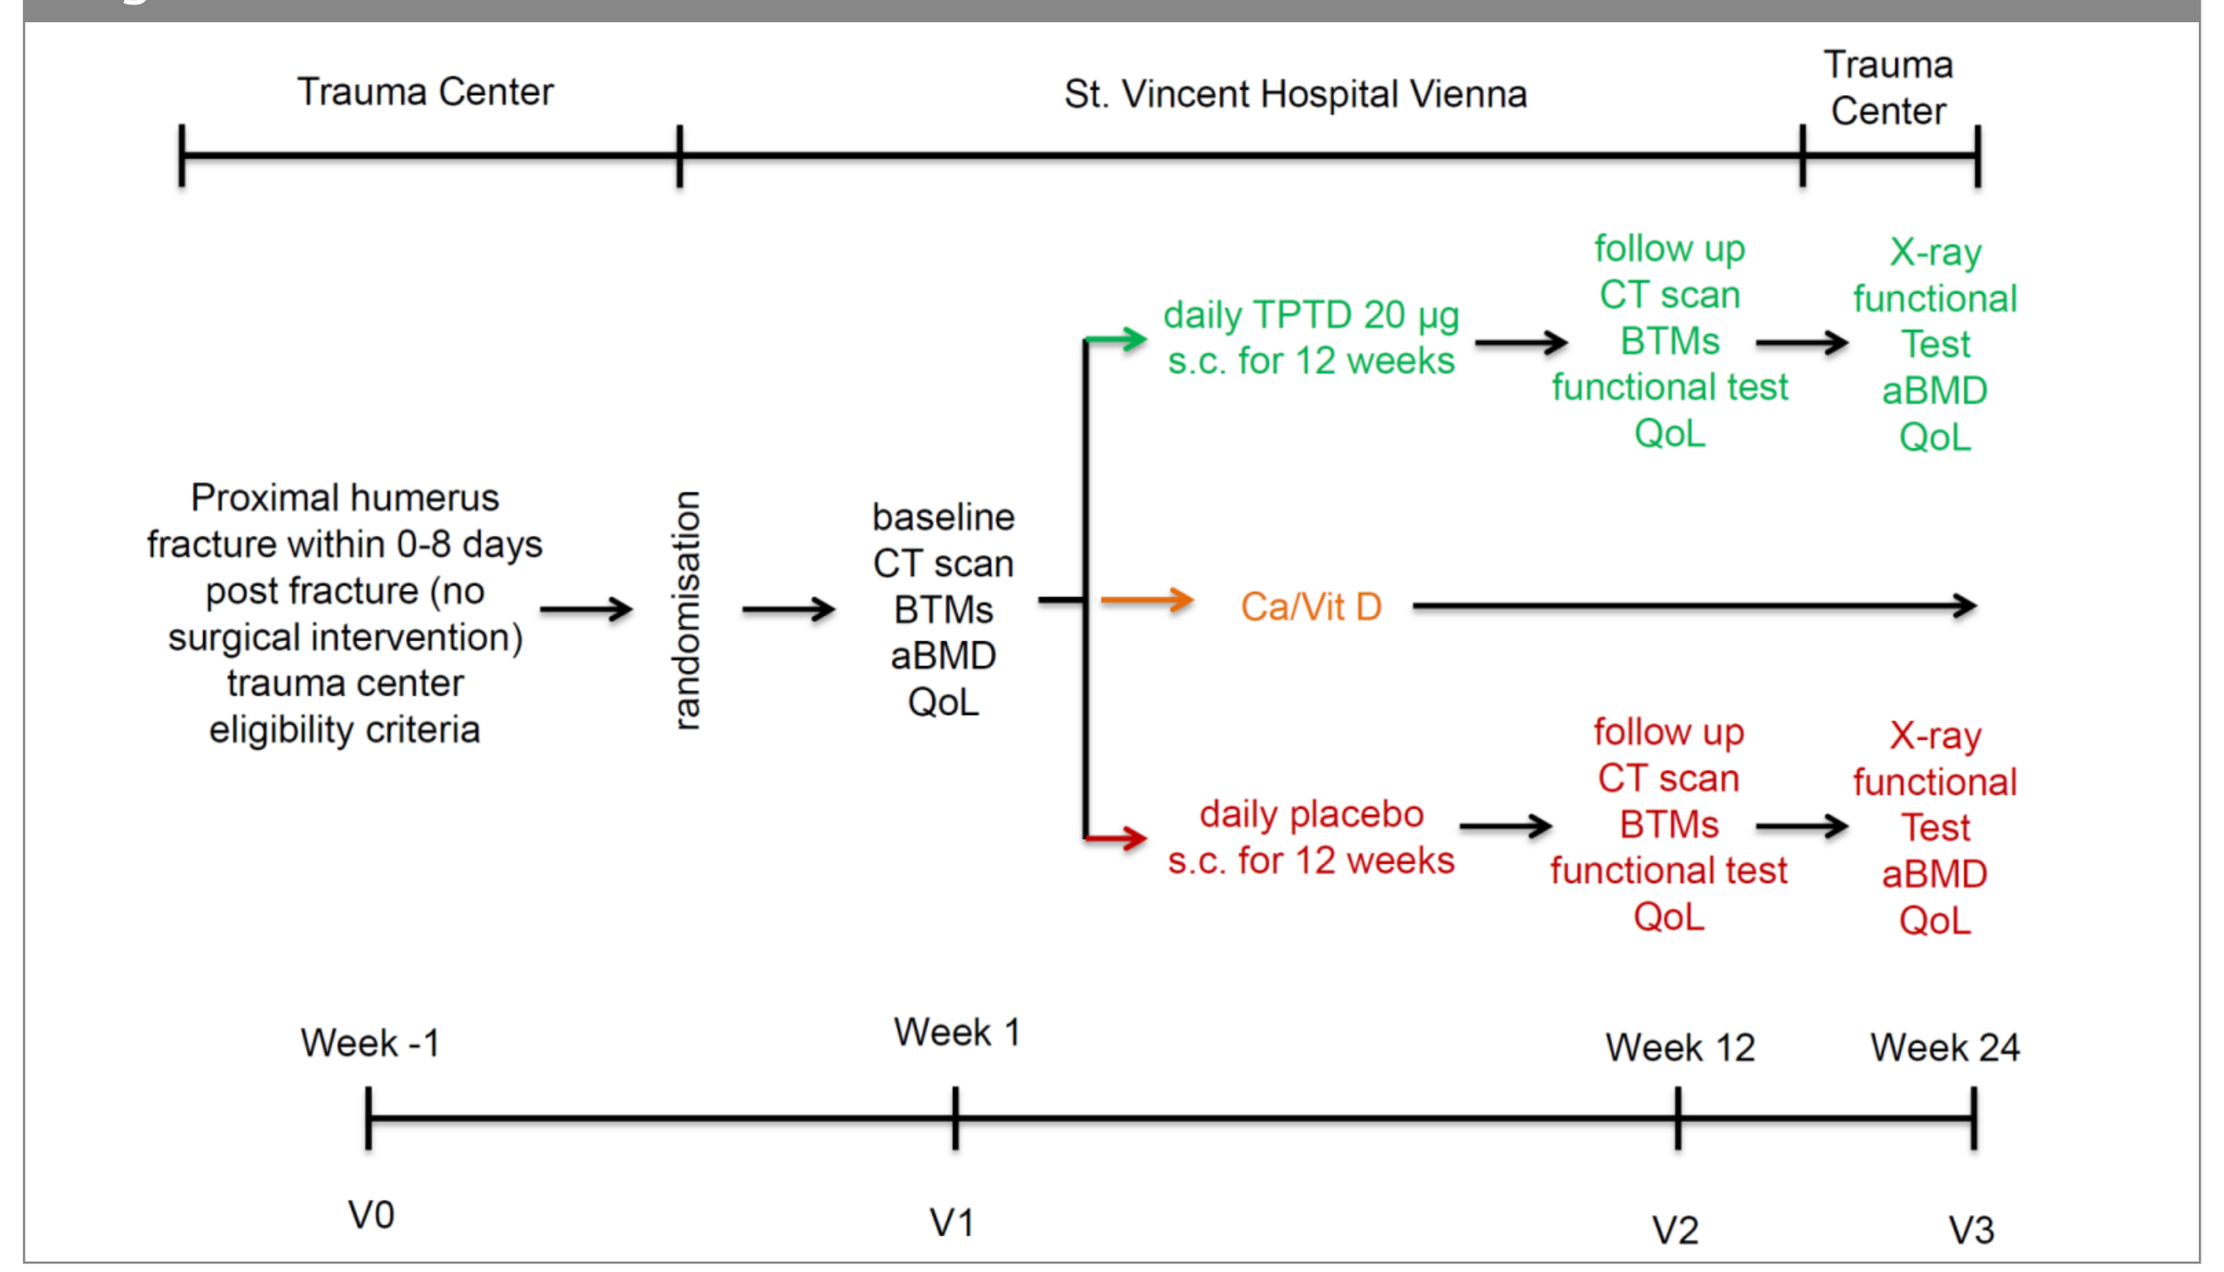
Trauma Center
St. Vincent Hospital Vienna
Trauma Center
Proximal humerus fracture within 0-8 days post fracture (no surgical intervention) trauma center eligibility criteria
randomisation
baseline CT scan BTMs aBMD QoL
daily TPTD 20 µg s.c. for 12 weeks
follow up CT scan BTMs functional test QoL
X-ray functional Test aBMD QoL
Ca/Vit D
daily placebo s.c. for 12 weeks
follow up CT scan BTMs functional test QoL
X-ray functional Test aBMD QoL
Week -1
Week 1
Week 12
Week 24
V0
V1
V2
V3

Figure 1: Study flow chart

Methods

This prospective double-blind placebo-controlled study investigated the effects of daily subcutaneous 20 µg TPTD induced changes in 3D bone structure over a 12-weeks treatment period.

Untreated postmenopausal women and men with a recent non-surgically treated proximal humerus fracture (≤ 8 days) and without any comorbidity known to affect bone metabolism participated. To quantify localized changes over time, affine and subsequent non-rigid registration of baseline and follow-up high-resolution CT scans (128-row scanner, Somatom Definition AS+, Siemens) were performed. Scans covered proximal and distal humerus, with the arm constrained. At baseline one region of interest at the fracture was annotated for each patient. This region was computationally mapped to the same location in the F/U scan, and average Hounsfield unit values (HU) were extracted in the same region.

The II° objectives included changes of baseline adjusted values of bone turnover markers (BTM) of formation (P1NP) and resorption (CTX), areal BMD (aBMD: iDXA, DXL) at spine, hip and calcaneus, changes in upper arm mobility (Constant Shoulder Score), pain (VAS) and SF-36 quality of life (QoL) scores.

Results:

Fifty-three of 82 screened patients (42 females – mean age 74.1 ± 11.0 years; 11 males; mean age 69.3 ± 8.6 years) were randomized either to TPTD or placebo, with 13 patients excluded (surgery, withdrawal). Forty patients finished the trial.

In the fracture regions of the TPTD patients superior increases of HU values from baseline to follow up (average delta, +176.1; p<0.001 vs. placebo +125.2; p=0.007) were observed.

Simultaneously TPTD induced increases of P1NP levels by 59 µg/L, and CTX by 0.31 ng/mL (p<0.005 for both).

Areal BMD improved at spine and hip (p<0.05 for both).

QoL, VAS and mobility changes were more intense with TPTD treatment compared to placebo (p<0.05 for all) – data not shown

Parameter	Effect (∆ change) TPTD – placebo (baseline adjustment)	Confidence interval low	Confidence interval high	P-value
DXA parameters				
L1-L4 (g/cm2)	0.030	0.002	0.058	**0.033**
Femoral neck (g/cm2)	0.021	0.001	0.041	**0.046**
Total hip (g/cm2)	0.023	0.007	0.039	**0.006**
FSI	-0.048	-0.420	0.324	0.795
CSMI (mm4)	0.588	-0.167	1.343	0.122
CSA (mm2)	6.258	1.985	10.532	**0.005**
Radius lower 1/3 (g/ cm2)	0.044	0.095	0.007	0.091
Calcaneus (left) (g/cm2)	0.015	0.030	0.001	**0.041**
Serum values				
Calcium (mmol/L)	-0.03	-0.09	0.03	0.345
Phosphate (mmol/L)	-0.03	-0.12	0.06	0.489
iPTH (pg/mL)	-6.68	-19.53	6.15	0.296
25-OH vitamin D3 (ng/mL)	-1.19	-7.63	5.24	0.708
CTX (ng/mL)	0.31	0.134	0.49	**0.001**
P1NP (µg/L)	59.03	24.60	93.46	**0.002**

Table 1: TPTD, teriparatide; DXA, dual energy x-ray absorptiometry; L1-L4, lumbar spine vertebrae 1-4; FSI, femur strengh index; CSMI, cross sectional moment of inertia; CSA, cross sectional area; iPTH, intact parathyroid hormone; CTX, serum type 1 collagen cross-linked C-telopeptide; P1NP, intact amino terminal propeptide of type I procollagen; significant p-values are bold

Figure 2

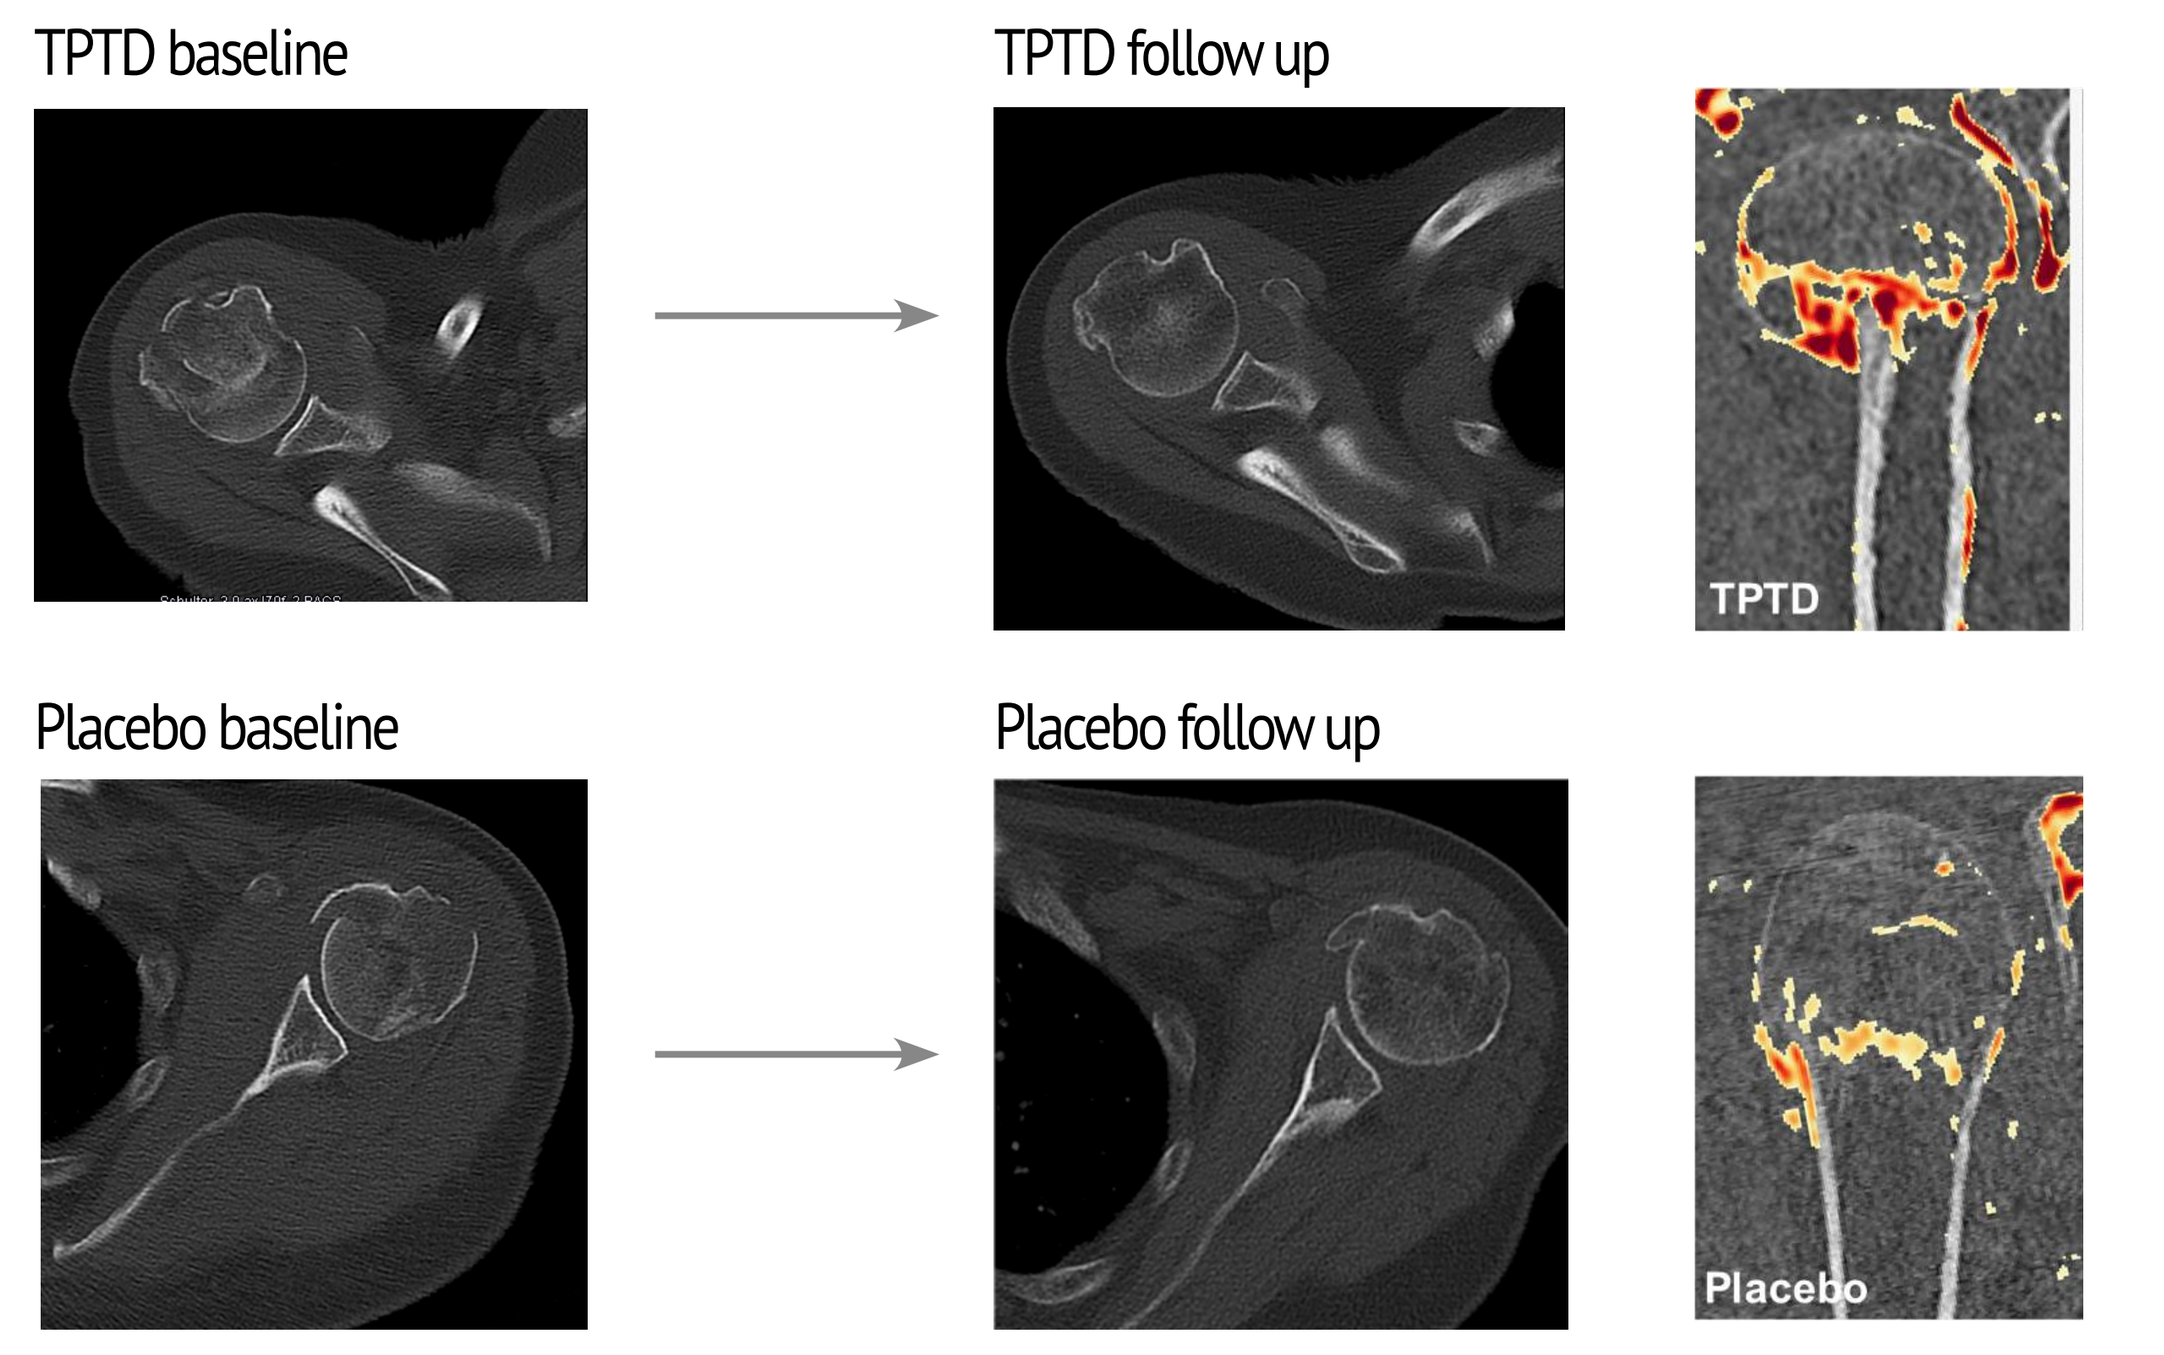
TPTD baseline
TPTD follow up
TPTD
Placebo baseline
Placebo follow up
Placebo

Figure 2: CT-scans at baseline and follow up (3D reconstruction – red areas inidcate increases of hounsfield [HU] units)

Adverse Events:

The overall safety profile was favorable. All reported adverse events were mild to moderate and predominantly occurred in the placebo group. No serious adverse events occurred. Three patients reported constipation, one patient transient pruritus, one patient with nausea, one patient decompensation of preexisting mild cardiac failure and one patient reported an increase of arterial hypertension. Only two patients of the placebo group transiently stopped study medication due to adverse event for less than 2 weeks.

Constipation: 2x placebo, 1x TPTD
Pruritus: 1x placebo
Nausea: 1x placebo (paused for 2 weeks)
Cardiac decompensation: 1x placebo (paused for 2 weeks)
Mild increase of hypertension: 1x TPTD

Conclusion:

After a proximal humerus fracture an early initiation of a daily 12-week administration of TPTD exerts favorable effects at the fracture region through rapid increases of BTM of formation. Additionally these patients benefit from enhanced upper arm mobility, a robust pain reduction and an improvement of their QoL after TPTD.

Funding source: Investigator initiated study – Eli Lilly and Company IIT B3D-OE-O042

Registrations:
EudraCT: 2014-002154-39:
Clinical Trials: NCT01739855

VINforce

ACADEMIC TEACHING HOSPITAL MEDICAL UNIVERSITY VIENNA

MEDICAL UNIVERSITY OF VIENNA